LIFE SCIENCES, GENOMICS AND BIOTECHNOLOGY FOR HEALTH

P1804

Evaluation of RespiFinder Smart for the detection of respiratory viruses

K. Loens[1]*, F. Coenjaerts[2], E. Claas[3], H. Goossens[1], A. van Loon[2], M. Ieven[1] on behalf of GRACE WP3

[1] University of Antwerp, Antwerp, Belgium; [2] Universitair Medisch Centrum Utrecht, Utrecht, The Netherlands; [3] Leiden University Medical Centre

GRACE

Genomics to combat Resistance against Antibiotics in Community-acquired LRTI in Europe

OBJECTIVES

We investigated the viral aetiology in lower respiratory tract infections (LRTI) in the European GRACE primary care network (PCN) using mono- and small multiplex real-time nucleic acid amplification tests (NAATs). These had been shown to be more sensitive than some large multiplex (MX) assays on a proficiency panel, but are more time-consuming due to the large diversity of respiratory viruses. This study compares the performance of RespiFinder® Smart kit (RFS, PathoFinder), a new and convenient real-time MX assay, to in-house real-time PCRs on nasopharyngeal flocked swabs (NPFSs, COPAN, Brescia, Italy).

MATERIALS & METHODS

568 NPFSs (kindly provided by COPAN) were collected prospectively in 11/16 PCNs in 8 countries during three winter seasons from 1 October 2007 until 30 March 2010. They were sent to the local lab to be frozen until transport to the central lab in Antwerp for nucleic acid (NA) extraction by the NucliSens EasyMAG (bioMérieux). Aliquots of NA extracts were sent to LUMC and UMC-UTRECHT for detection of influenzaviruses (INF) A/B, parainfluenzavirus (PIV) 1-4, human rhinoviruses (HRV), human metapneumovirus (hMPV), respiratory syncytial virus (RSV), adenoviruses (HAdV), bocavirus (HBoV), and coronaviruses (HCoV) by in-house monoplex and small MX real-time PCR (1). The RFS kit was retrospectively applied in Antwerp. According to the instructions of the manufacturer. Sensitivity and specificity were calculated against in-house PCRs.

References

1. Loens K, van Loon AM, Coenjaerts F, van Aarle Y, Goossens H, Wallace P, Claas EJ, Ieven M; GRACE Study Group. 2012. Performance of different mono-and multiplex nucleic acid amplification tests on a multipathogen external quality assessment panel. J Clin Microbiol. 50(3):977-87.

Correspondence:

Greet Ieven, leader WP3, greet.ieven@uza.be
Katherine Loens, Katherine.loens@uza.be
Vaccine & Infectious Disease Institute (VAXINFECTIO)
University Hospital Antwerp, Wilrijkstraat 10, B-2650 Edegem, Belgium

This project was supported through Priority 1 (Life Sciences, Genomics and Biotechnology for Health) of European Union's FP6, Contract number: LSHM-CT-2005-518226

RESULTS

•331 and 349 respiratory viruses were detected by the real-time in-house PCRs and RFS, respectively. Using in-house PCRs, the following viruses were detected: HAdV (n=13), PIV1-4 (n=36), HRV (n=96), hMPV (n=36), RSV (n=42), hBoV (n=18), INF A/B (n=41), HCoV (n=48). Dual viral infections were detected in 12 and 26 samples by in-house PCR and RFS, respectively (Table 1).

•Sensitivities and specificities of RFS for the detection of the individual viruses as well as the overall sensitivity and specificity are shown in Table 2.

•In general, samples found negative by RFS and positive by the mono-PCRs tended to have a low viral load (based on Ct-value >37.5, Table 3) or represented dual infections.

Table 1. Dual viral infections

Virus 1	Virus 2	In-house PCR (n=12)	RFS (n=26)
HAdV	RSV	2	1
hMPV	HCoV OC43	1	2
hMPV	HCoV NL63	1	1
HRV(/EV)	HCoV NL63	1	1
HRV(/EV)	HCoV 229E	1	8
HRV(/EV)	HCoV OC43	1	0
HRV(/EV)	INF A	1	0
HRV(/EV)	hBoV	1	3
HRV(/EV)	RSV	0	1
HRV(/EV)	PIV	0	2
INF A	HCoV 229E	0	1
INF A	hMPV	0	2
PIV	INF A	1	0
PIV	HCoV 229E	1	1
RSV	HCoV 229E	1	1
RSV	ADE	1	0
RSV	HCoV HKU1	0	1
RSV	INF A	0	1

www.grace-lrti.org

CONCLUSIONS

•In-house PCRs were more sensitive than the RFS assay.

•Except for the detection of HadV, for which the sensitivity is rather low, RFS offers a valuable alternative to detect the presence of 17 viruses in a single tube.

Table 2. Sensitivities and specificities of the RespiFinder Smart compared to in-house PCRs

Virus	RFS+/in-house+	RFS+/in-house-	RFS-/in-house+	Sensitivity	Specificity
HAdV	6	2	7	46,2	98,6
PIV1-4	32	4	4	88,9	99,2
HRV/EV*	89	24	7	92,7	94,9
hMPV	33	3	3	91,7	99,4
RSV A/B	41	1	1	97,6	99,8
INF A/B	38	4	4	90,5	99,2
hBoV	15	2	3	83,3	99,6
HCoV**	36	19	12	75,0	96.3
Overall	290	59	41	87,6	98,6

*The RFS detects both HRV and EV in contrast to the in-house PCR.
** In-house PCR does not detect HCoV HKU1, whereas it is included in the RFS.

Table 3. Ct-values of RFS-/in-house+ samples

Virus	Ct-values
HAdV	37,05; 32,63; 38,21; 37,43; 38,80; 37,20; 35,81
PIV1-4	39,55; 38,74; 37,05; 38,53
HRV	36,10; 39,50; 36,78; 36,28; 29,64; 25,62; 34,78
hMPV	30,24; 33,19; 34,56
RSV	40,23
INF A/B	29,28; 39,06; 39,87; 40,32
hBoV	36,31; 37,46; 35,99
hCoV	39,32; 40,37; 35,31; 40,91; 40,44; 38,39; 39,97; 35,84; 39,53; 41,31; 37,85; 17,60